Protein therapeutics for cytoplasmic targets

Paulus Nyman[1], **Tuomas Huovinen**[1], **Ralf Paul**[2], and **Bryce Nelson**[2]

[1.] Department of Life Technologies, University of Turku,Turku, Finland

[2.] Protein and Antibody Engineering, Orion Corporation, Turku, Finland

Motivation

Antibody-based therapeutics have already proven the capabilities treating various diseases. Formation of disulfide bonds requires oxidative conditions and is not ideal when targeting intracellular proteins. Alternative binder proteins have been researched to tackle the challenges. DARPins (designed ankyrin repeat proteins) are small binder proteins. They stay functional even in cell cytoplasm. Other benefits are easy production in bacterial cells and good stability. These make DARPins an interesting research topic for therapeutic use.

DARPin library

A DARPin library has been constructed in Turku University by randomizing variable amino acids in DARPins. As a result, the library has up to 2.4 x10^{11} binders with different binding properties.

Biopanning

From the library, DARPins were selected against two undisclosed target proteins, X and Y using phage display. Bacteriophage contains DNA and expresses the corresponding DARPin at the surface. Only the phages expressing DARPin that binds to the provided target protein stay on immobilized target protein. Bacteria cells are infected with with phages eluted from the panning reaction. From cells, DNA can be extracted, or new phage clones can be produced. Panning rounds with and without detergent TWEEN were performed.

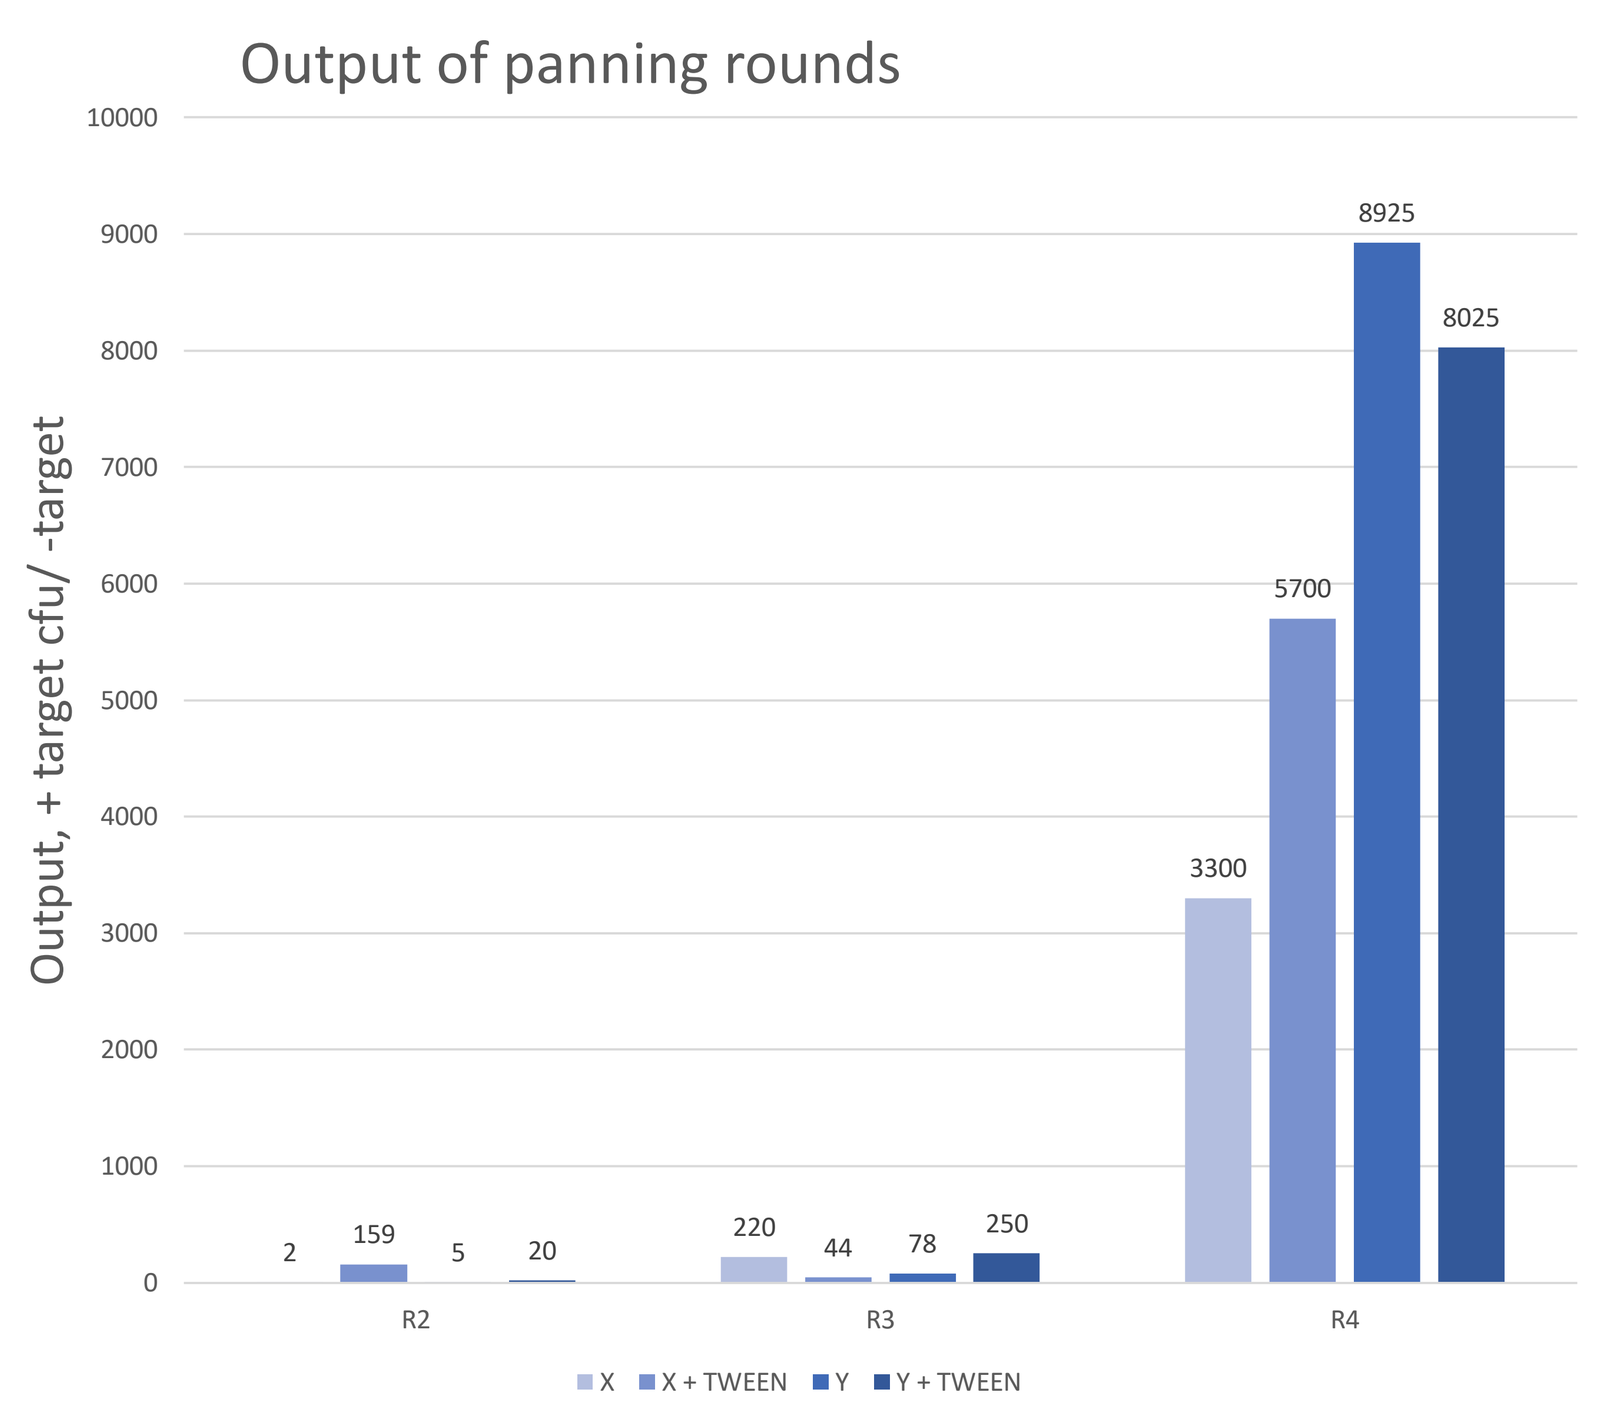
Output of panning rounds
Output, + target cfu/ -target
10000
9000
8000
7000
6000
5000
4000
3000
2000
1000
0
2
159
5
20
220
44
78
250
3300
5700
8925
8025
R2
R3
R4
X
X + TWEEN
Y
Y + TWEEN

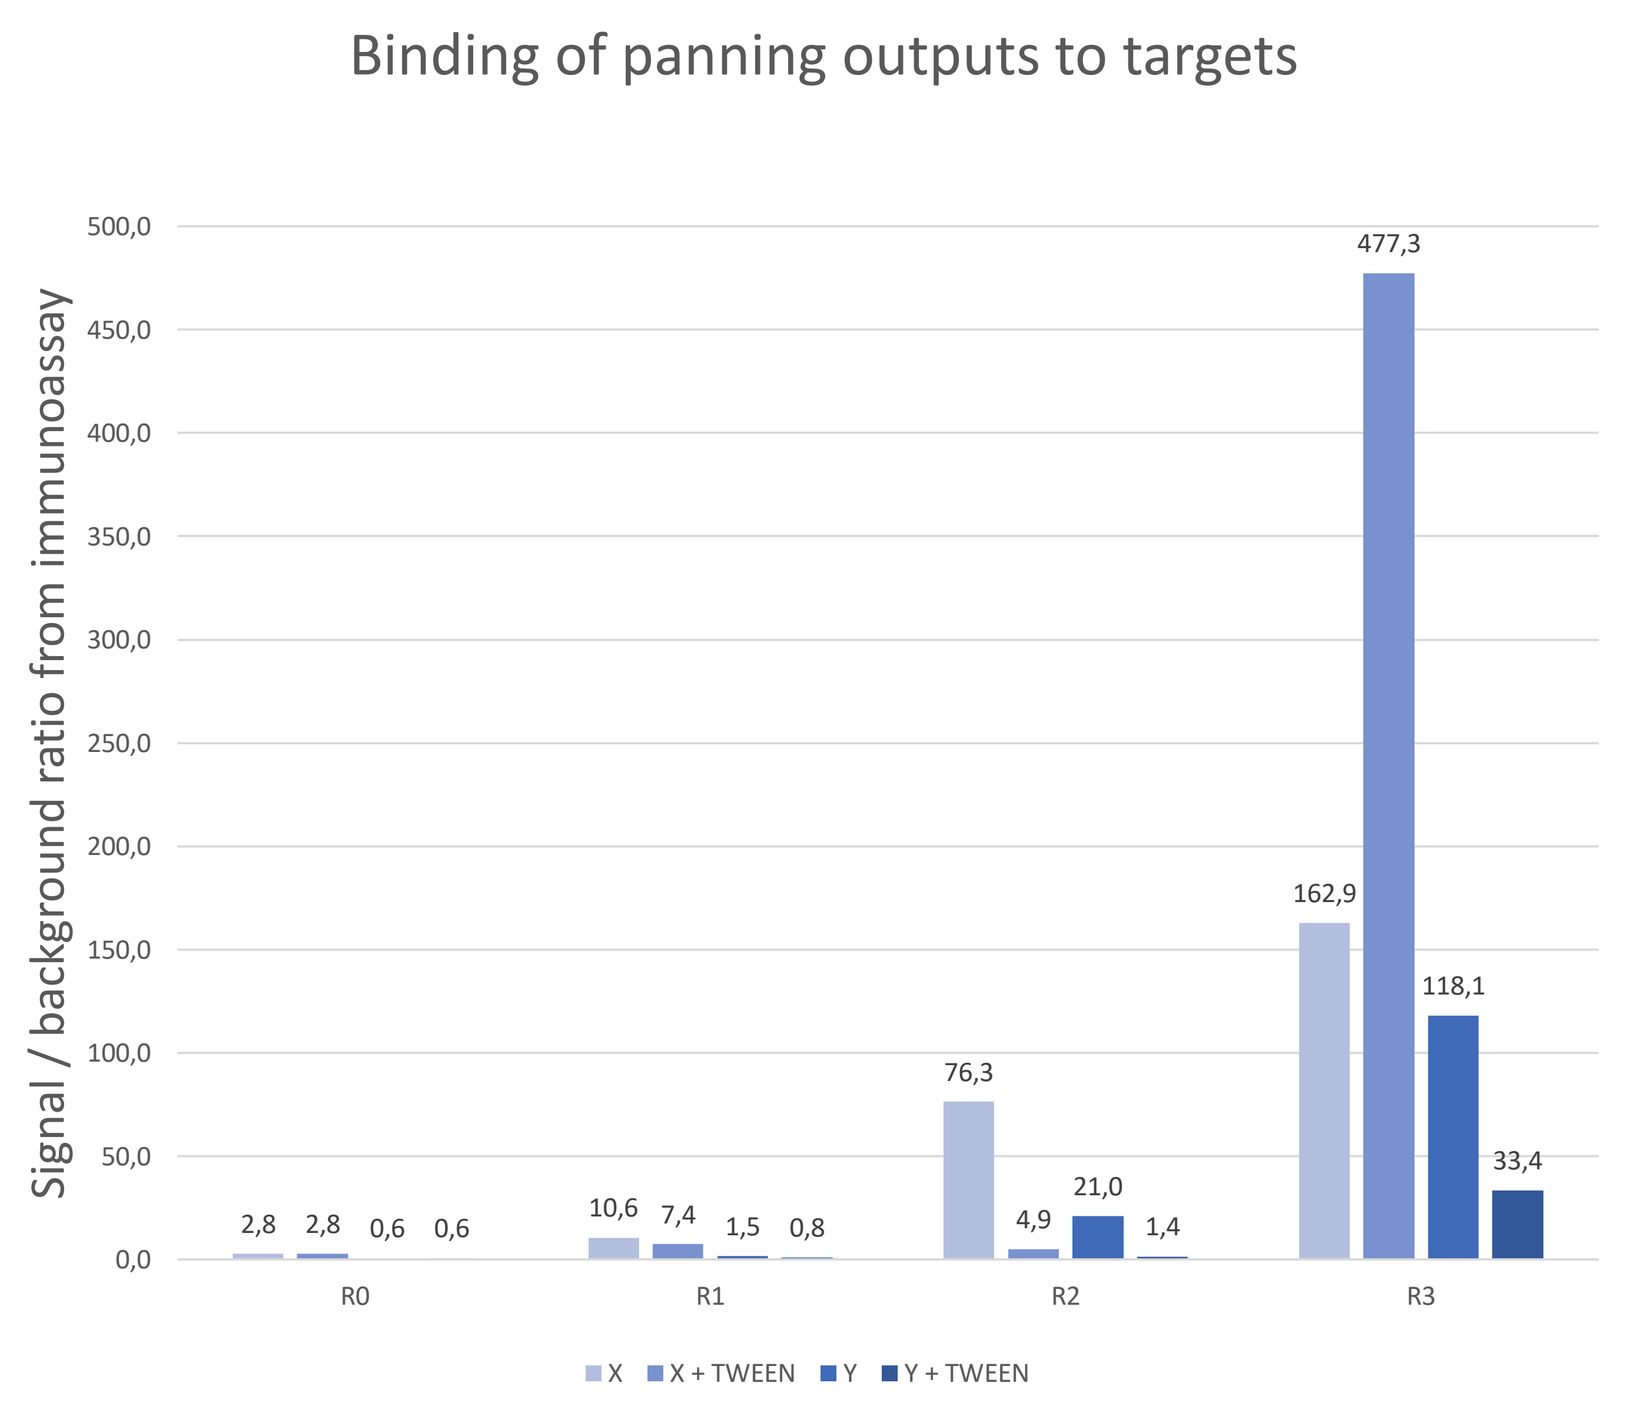
Binding of panning outputs to targets
Signal / background ratio from immunoassay
500,0
450,0
400,0
350,0
300,0
250,0
200,0
150,0
100,0
50,0
0,0
2,8
2,8
0,6
0,6
10,6
7,4
1,5
0,8
76,3
4,9
21,0
1,4
162,9
477,3
118,1
33,4
R0
R1
R2
R3
X
X + TWEEN
Y
Y + TWEEN

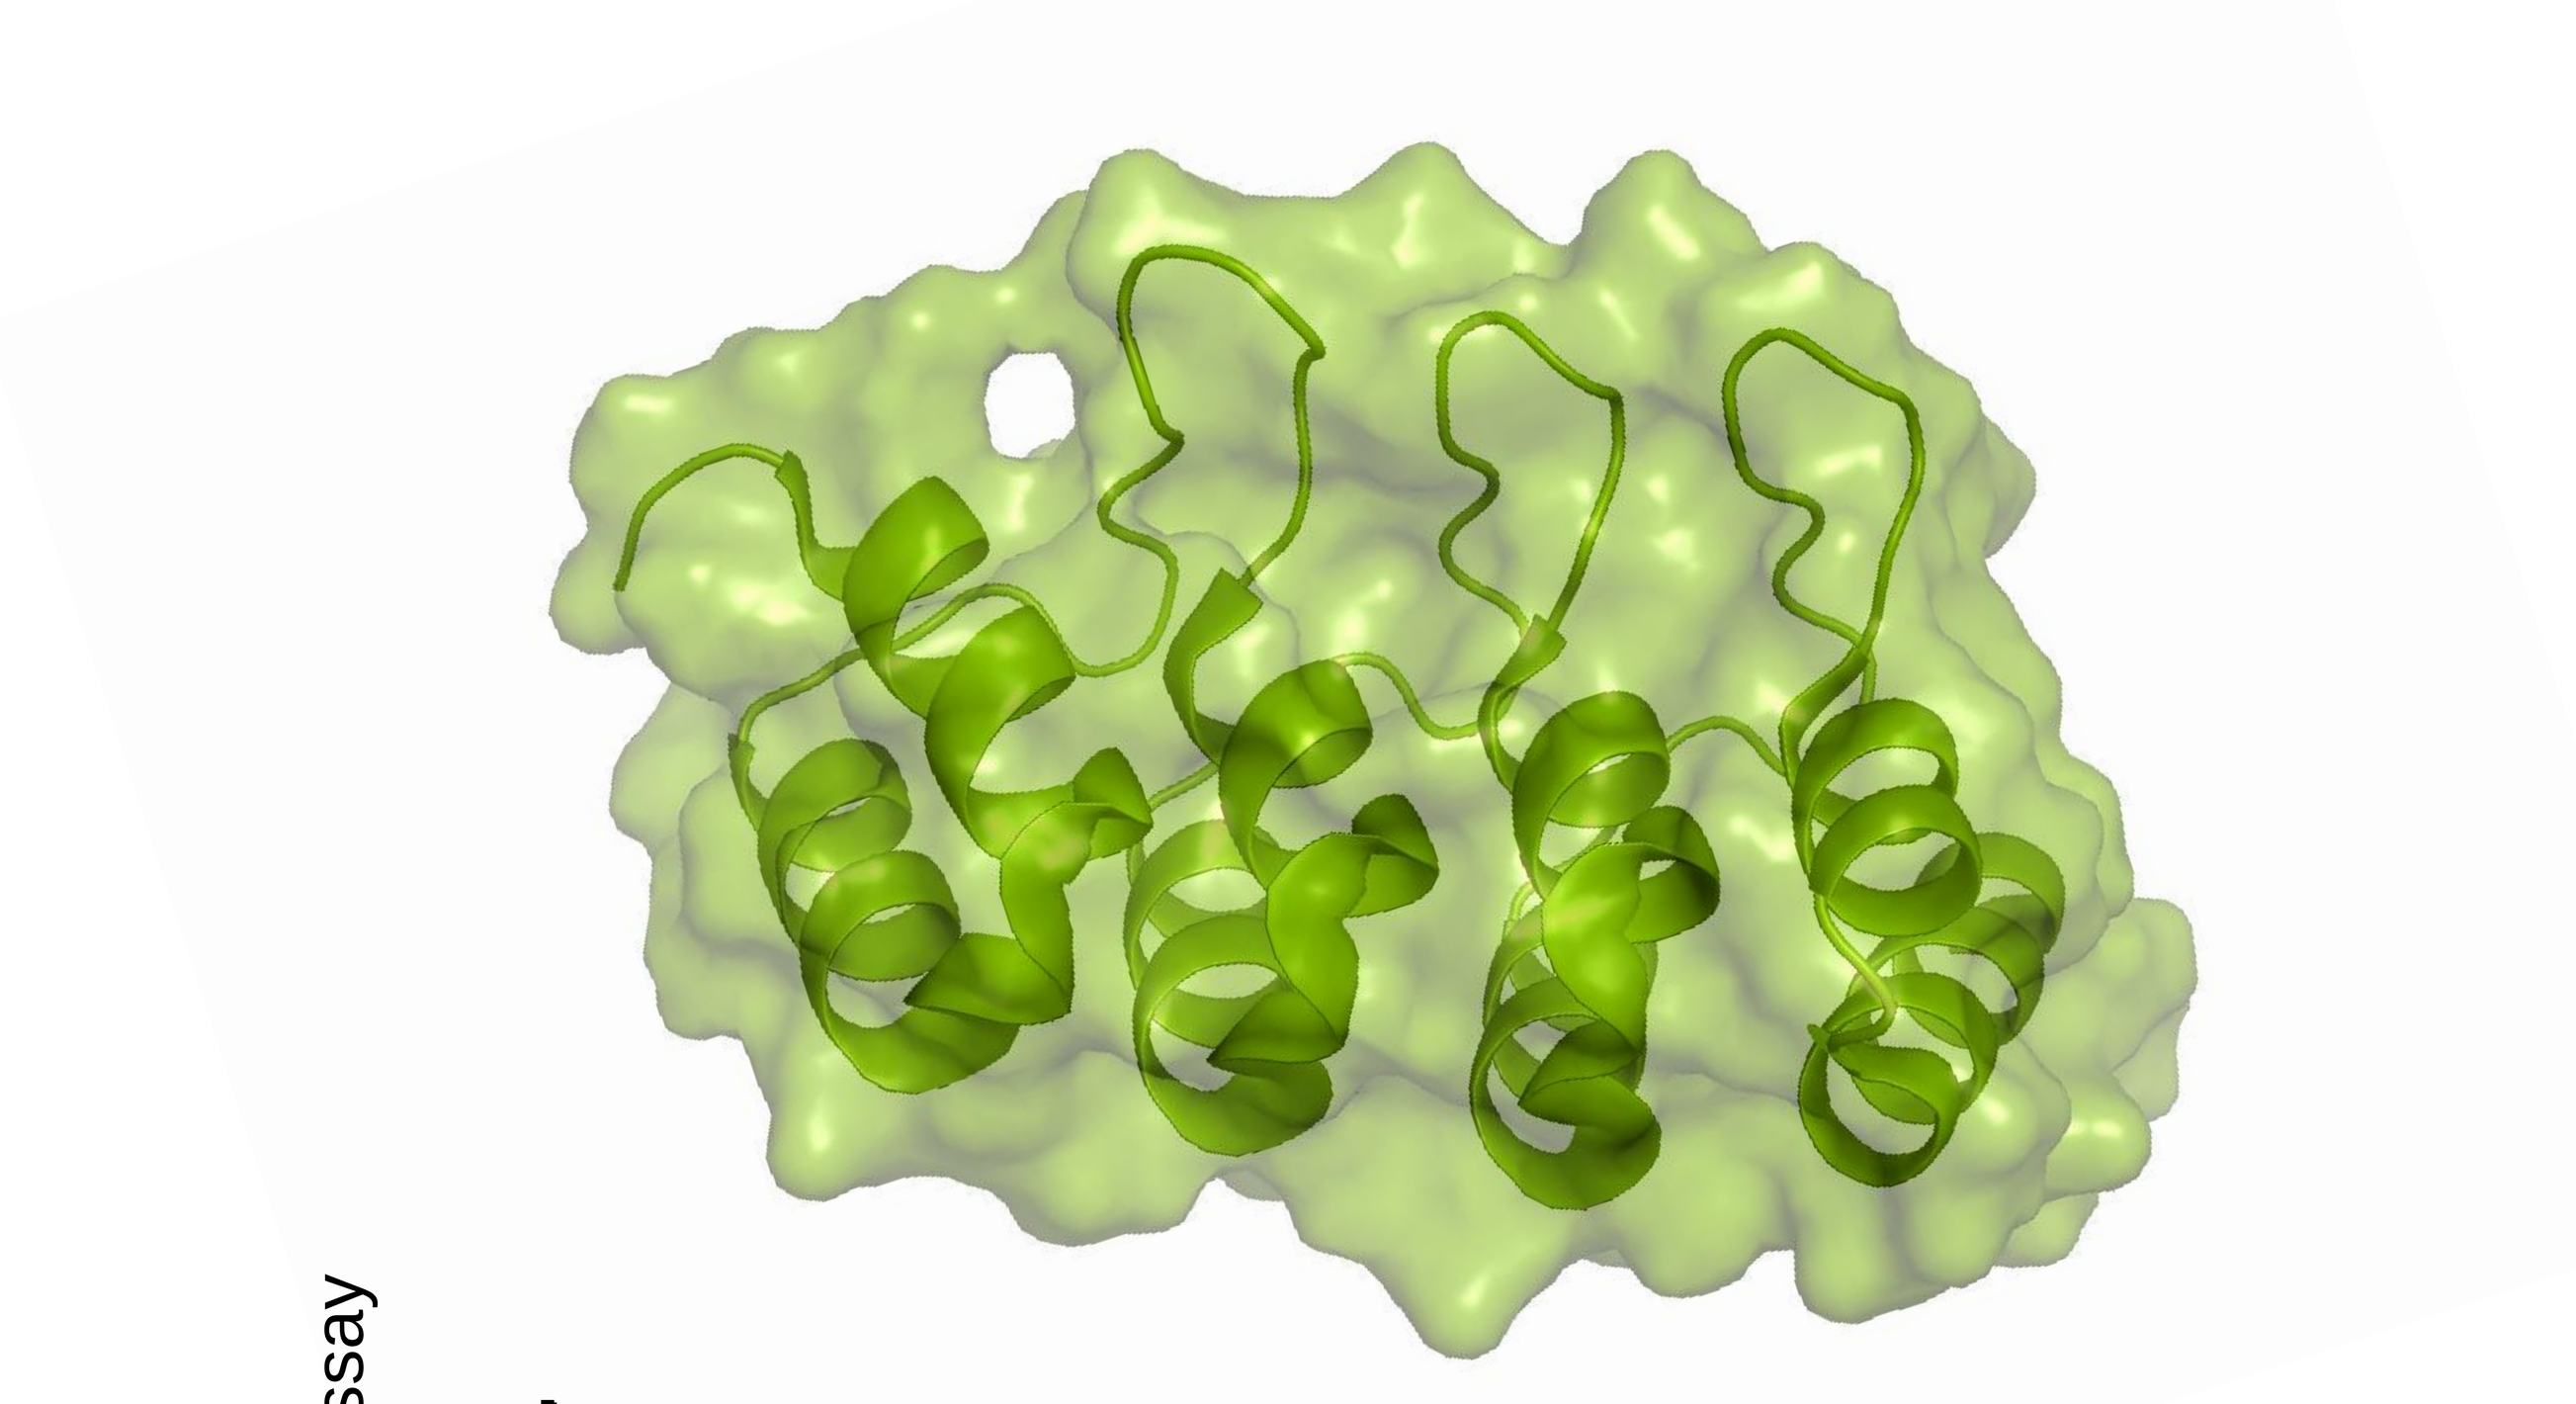

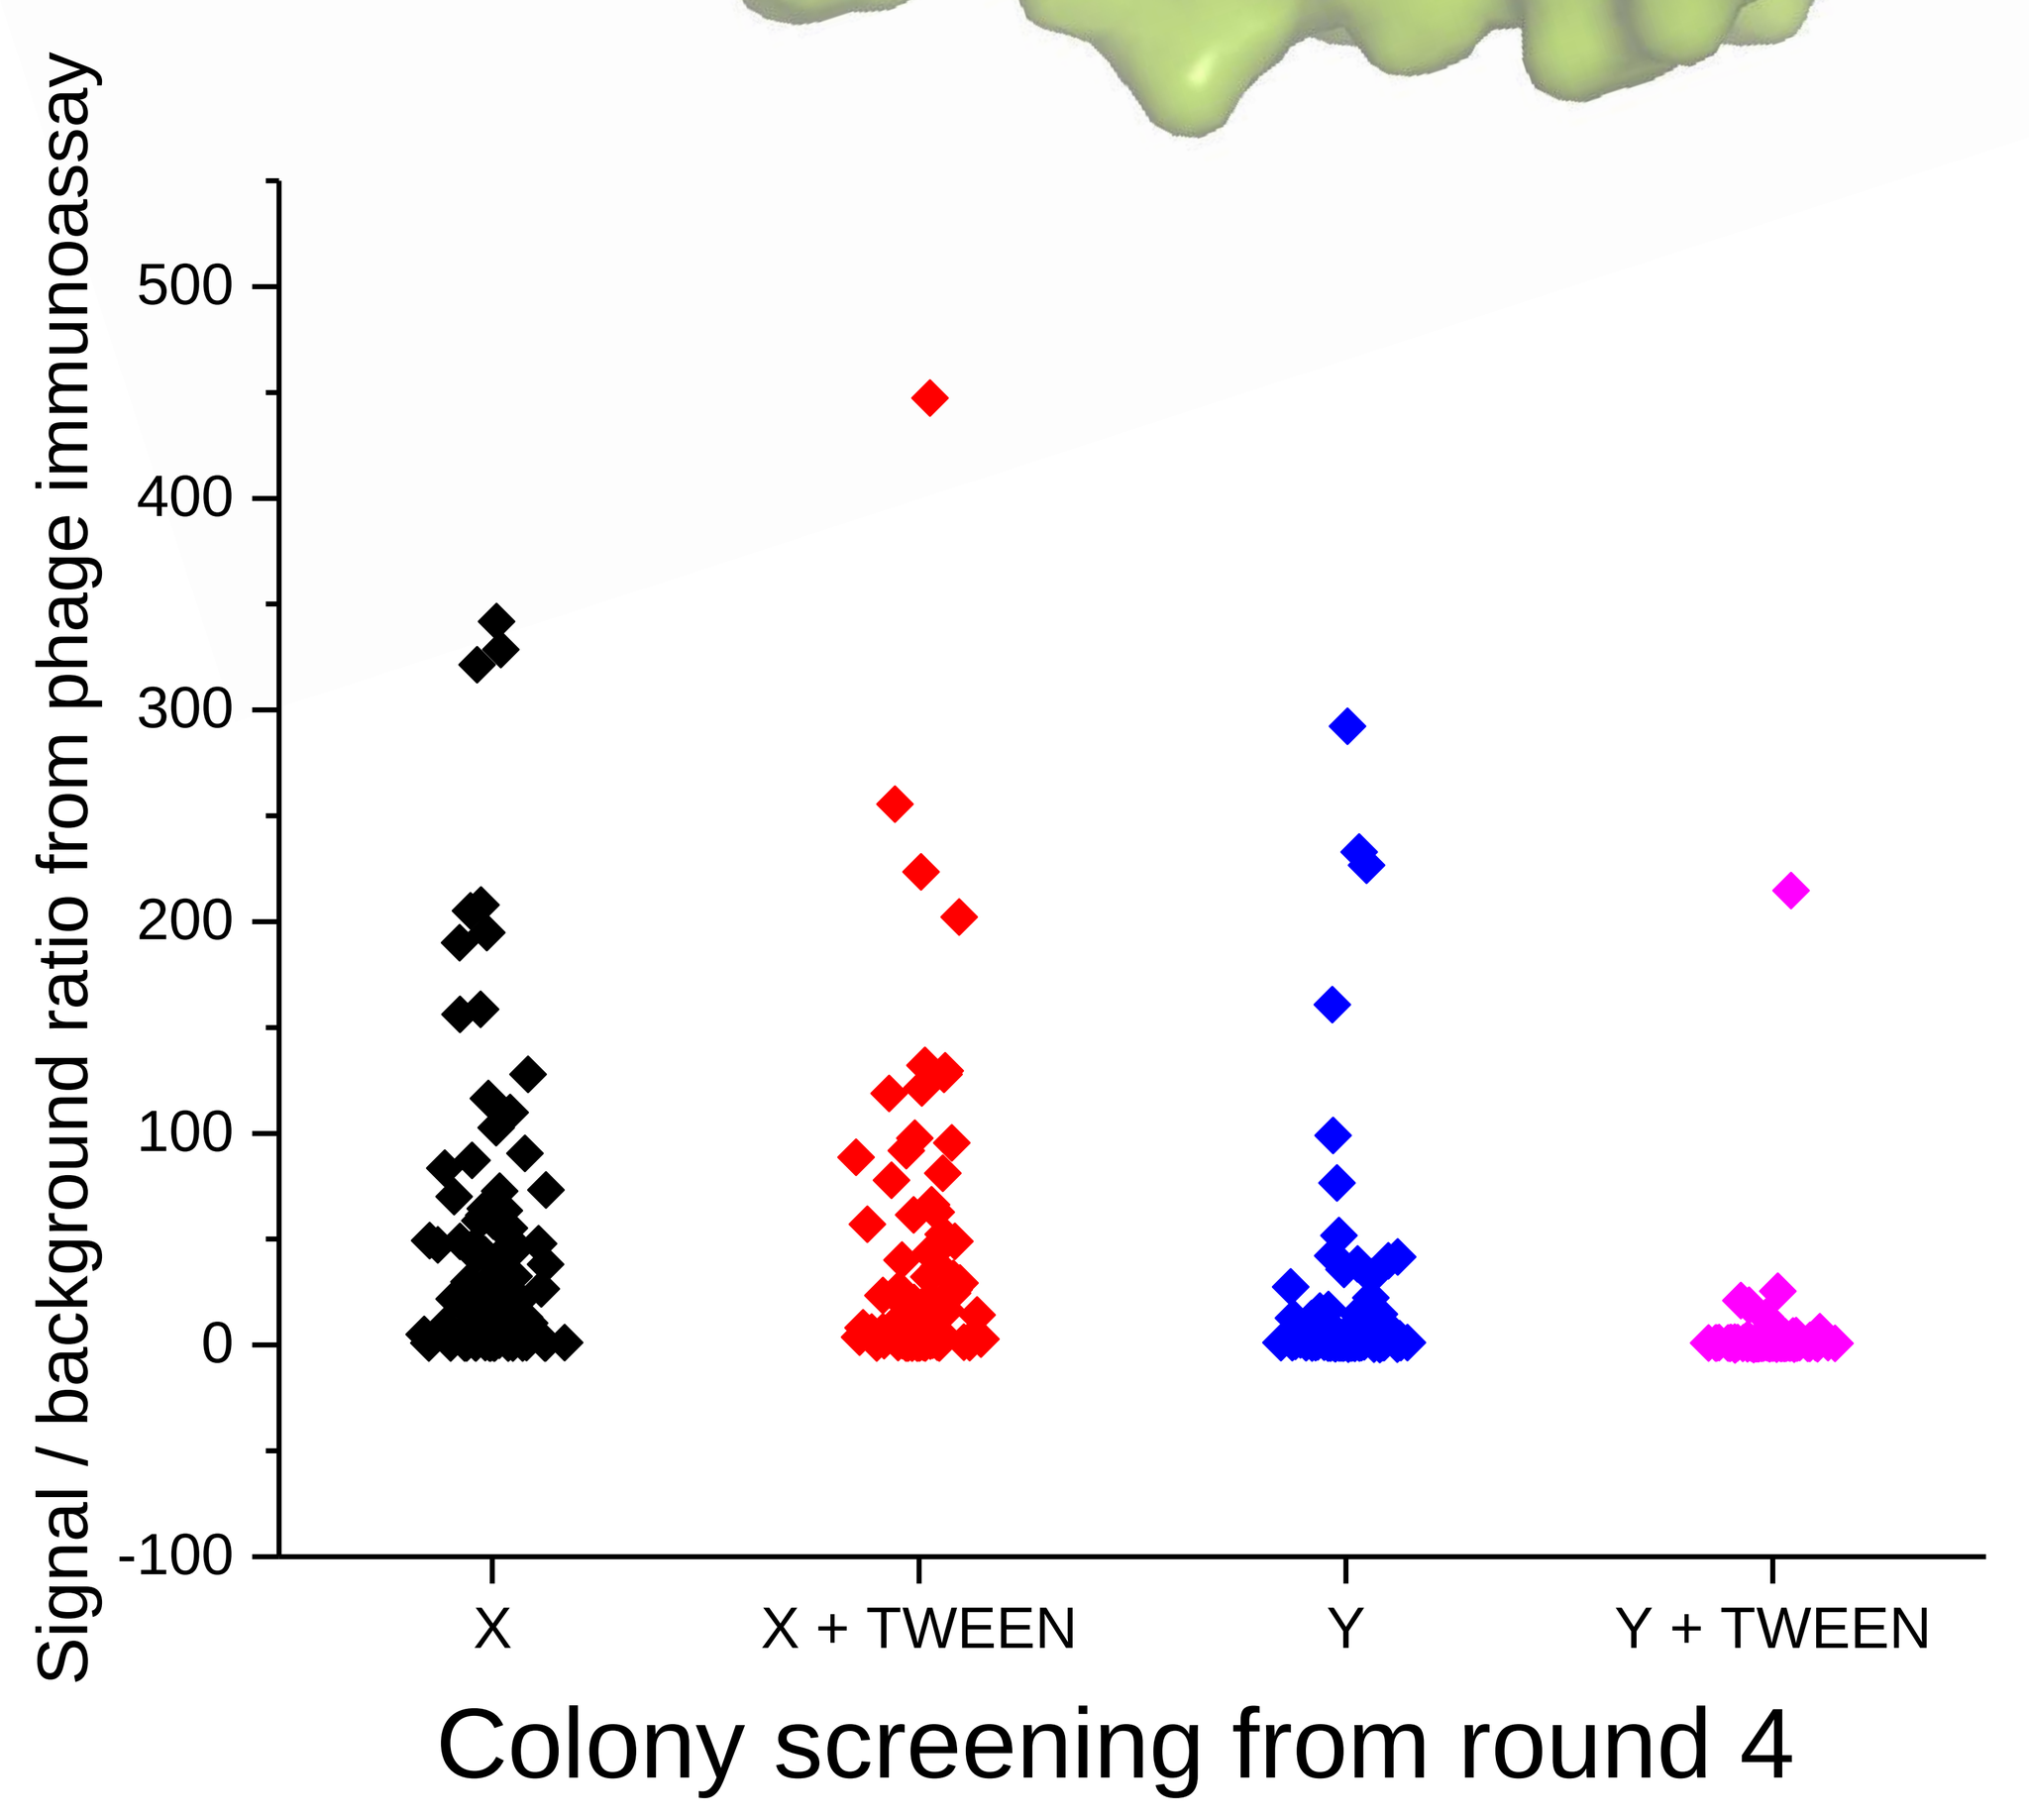
Signal / background ratio from phage immunoassay
500
400
300
200
100
0
-100
X
X + TWEEN
Y
Y + TWEEN
Colony screening from round 4

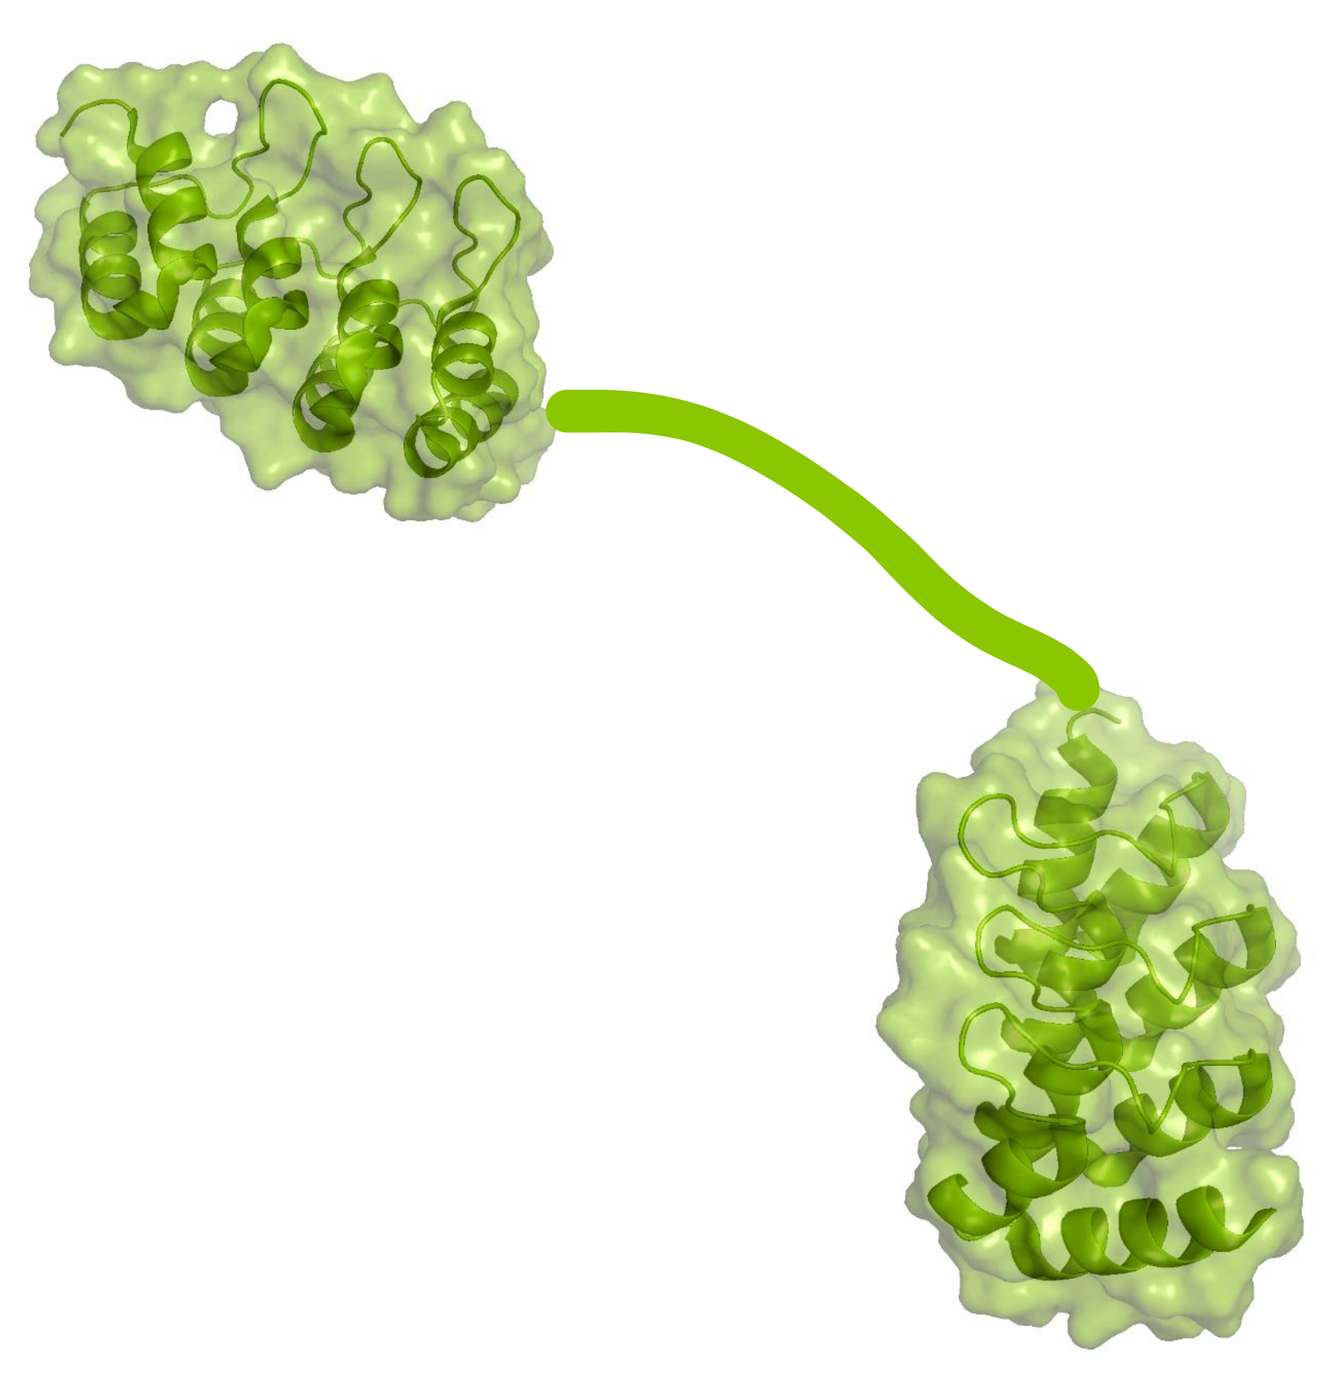

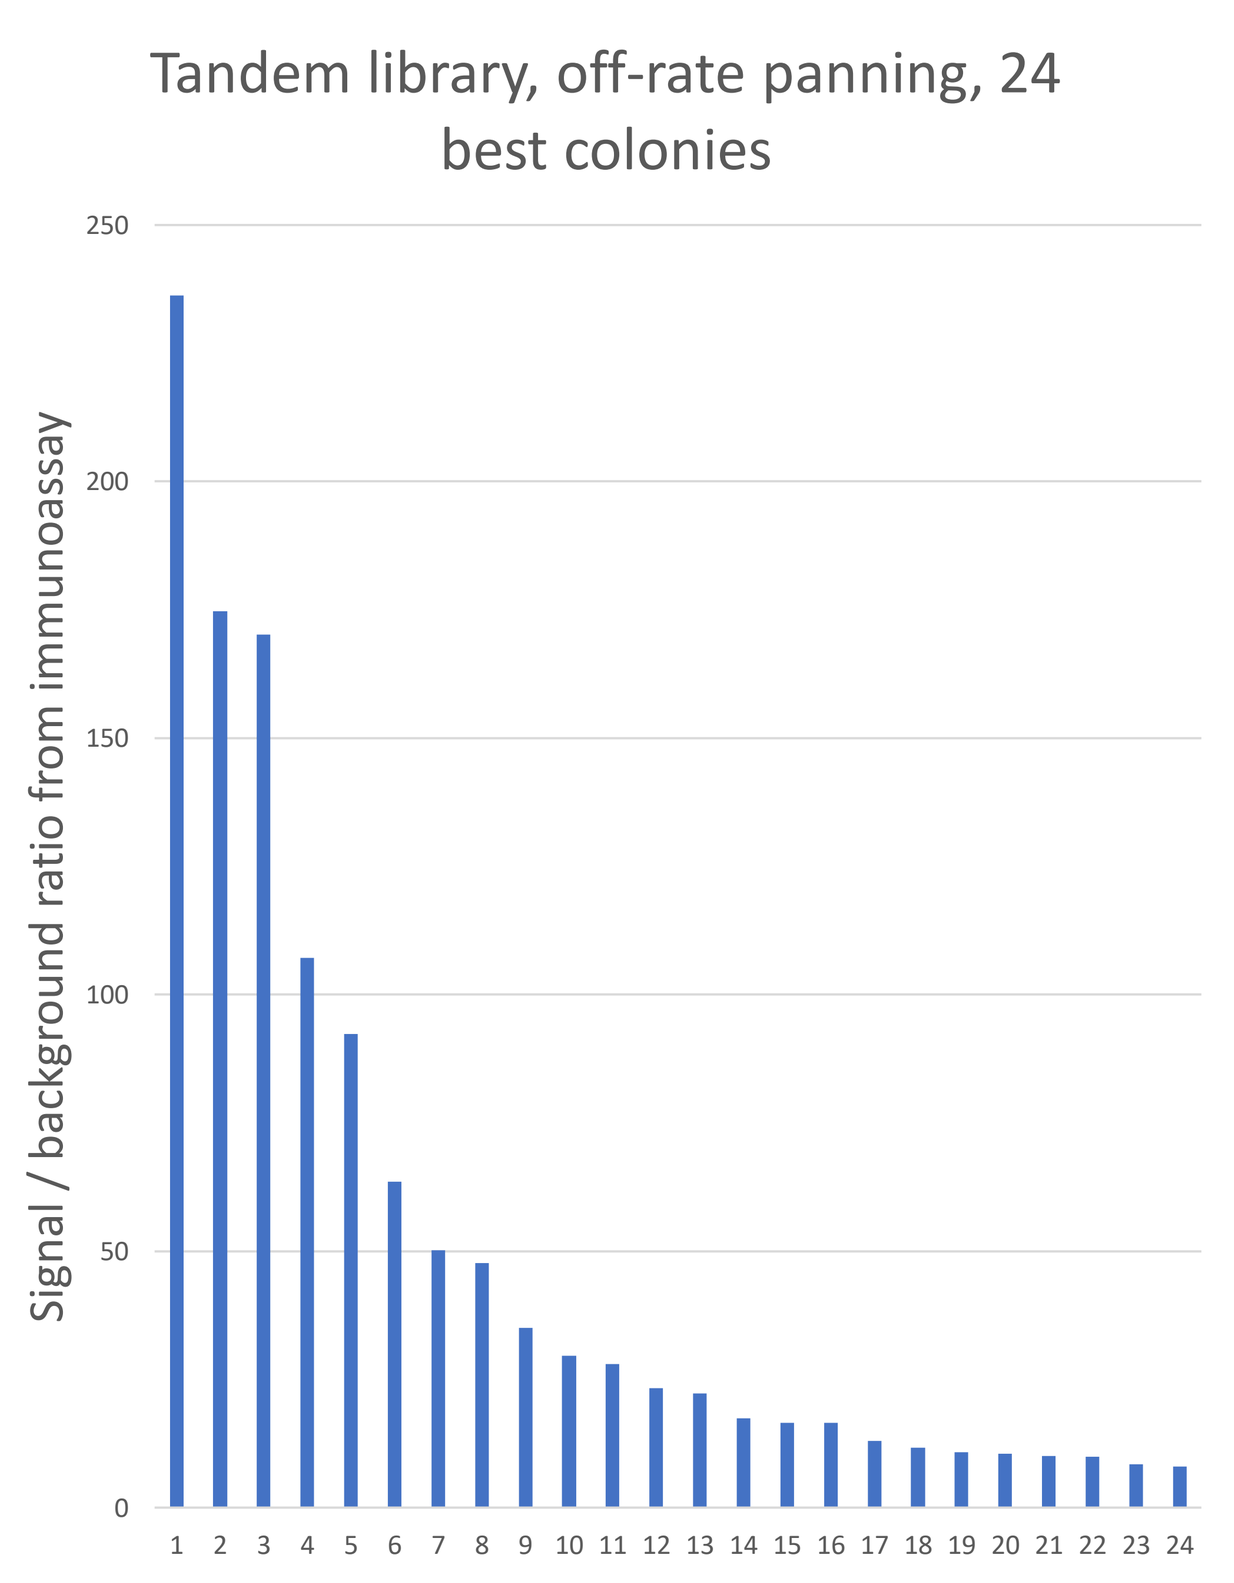
Tandem library, off-rate panning, 24 best colonies
Signal / background ratio from immunoassay
250
200
150
100
50
0
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24

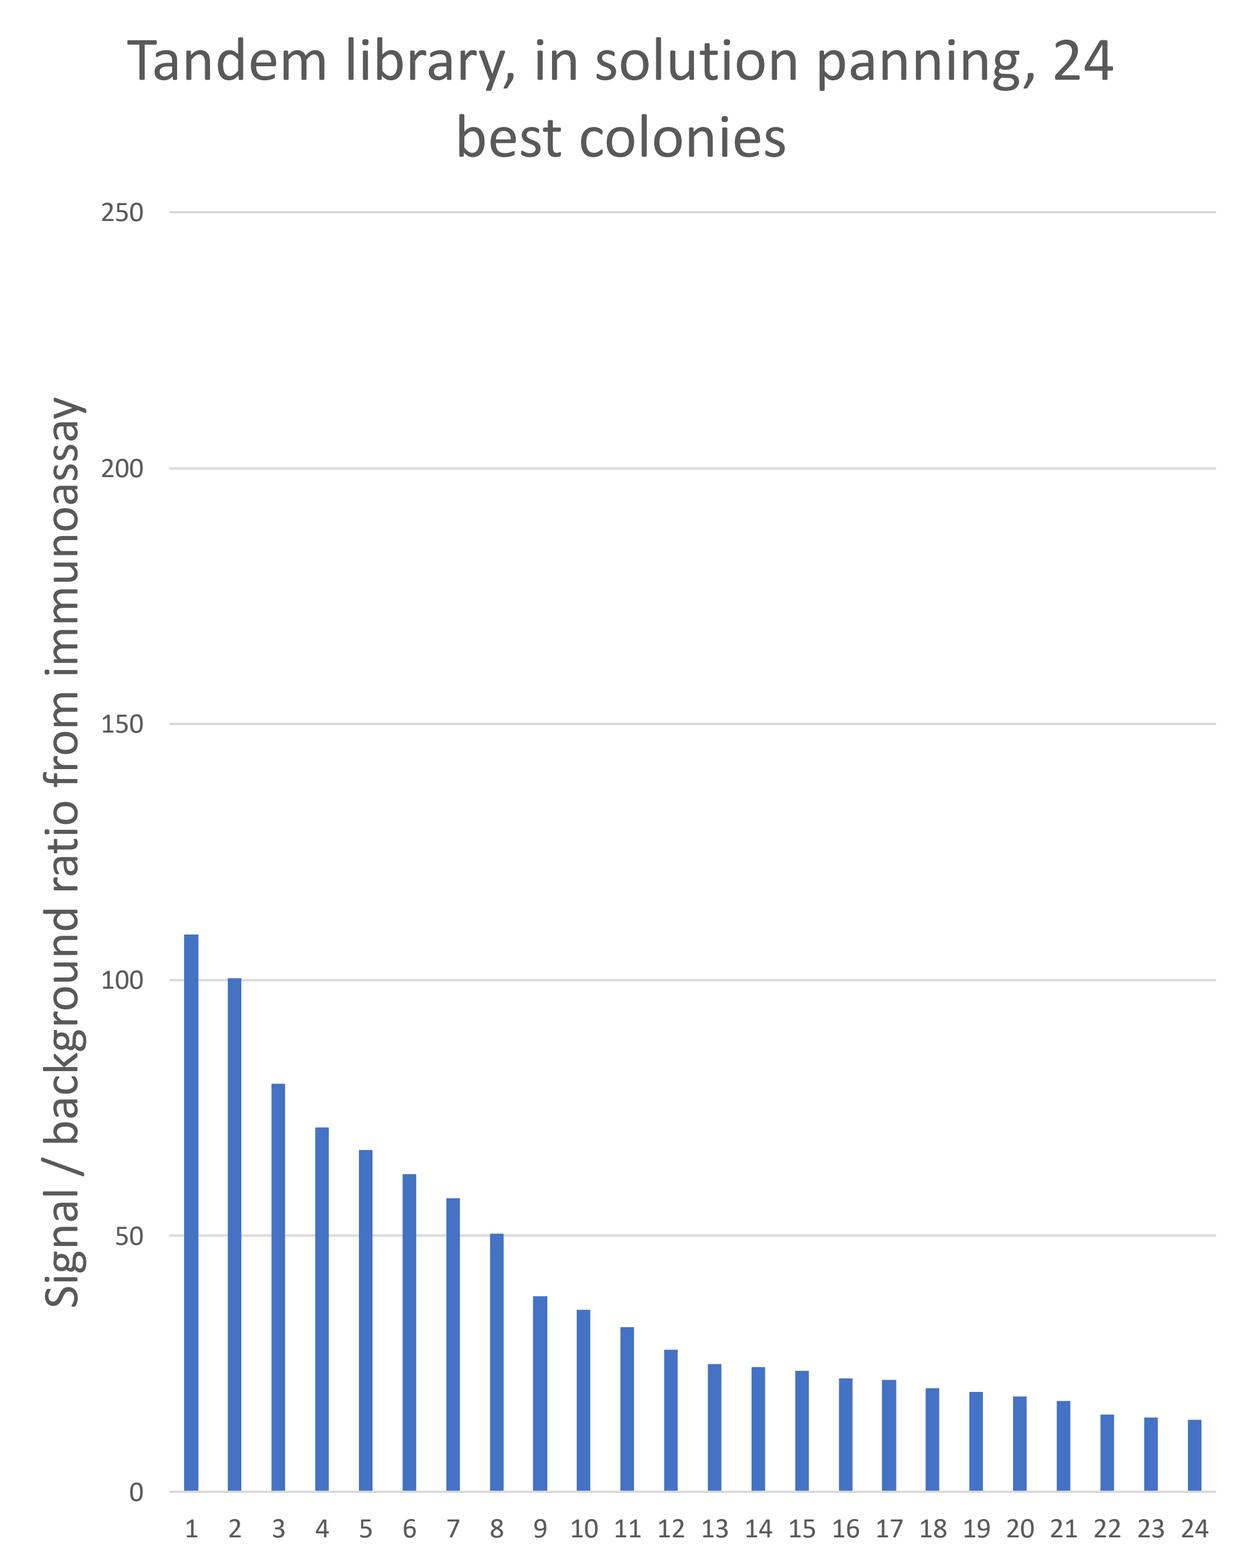
Tandem library, in solution panning, 24 best colonies
Signal / background ratio from immunoassay
250
200
150
100
50
0
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24

DARPin tandems

Bivalent binding has advantage over monovalent binding. For tandem library, DARPins were combined with linkers of variable lengths. The library was panned with and without off-rate method, by adding competitive target antigen after binding to biotinylated target.

Conclusions

Binders were found against both targets with both approaches. Off-rate panning was more efficient method for extracting binders with high binding capacity. The work was a proof of concept for using the DARPin library against cytoplasmic target proteins.